Evaluation of Implantable Cytokine Factories for Eradication of Malignant Pleural Mesothelioma (MPM) Tumors in Mice with Safe and Predictable Dosing in Non-human Primates

Amanda M. Nash[1,*], Samira Aghlara-Fotovat[1,*], Andrea Hernandez[1], Bertha Castillo[1], Aarthi Pugazenthi[2], Peter D. Rios[3], Sofia Ghani[3], Ira Joshi[3], Douglas Isa[3], Chunyu Xu[4], Rahul A. Sheth[5], Weiyi Peng[4], Jose Oberholzer[3], Amir A. Jazaeri[6], Hee-Jin Jang[2], Bryan M. Burt[2], Hyun-Sung Lee[2], Ravi K. Ghanta[2], and Omid Veiseh[1,#].

1Department of Bioengineering, Rice University Houston, TX, 2Michael E. DeBakey Department of Surgery, Baylor College of Medicine, Houston, TX, 3CellTrans, Inc., Chicago, IL, 4Department of Biology and Biochemistry, The University of Houston, Houston, TX, 5Department of Interventional Radiology, The University of Texas MD Anderson Cancer Center, Houston, TX, 6Department of Gynecologic Oncology and Reproductive Medicine, The University of Texas MD Anderson Cancer Center, Houston, TX, *These authors contributed equally

Background

Pro-inflammatory cytokines have been approved as a cancer immunotherapy for treatment of metastatic melanoma and renal carcinoma for over 30 years. However, widespread use of cytokine therapy in the clinic is limited by its short half-life in circulation and the associated toxicities that emerge as a result of high systemic exposure. To overcome these limitations, we developed a clinically translatable cytokine delivery platform, called cytokine factories, composed of genetically engineered epithelial cells encapsulated in biocompatible polymers. These cells are able to produce a wide range of natural cytokines (IL2, IL7, IL10, or IL12) and allow for controlled and predictable dosing in vivo. **Conclusions:** Our findings demonstrate the safety and efficacy of cytokine factories in preclinical animal models and provide rationale for future clinical testing for the treatment of metastatic peritoneal cancers in humans.

Tunable dose for personalized treatment

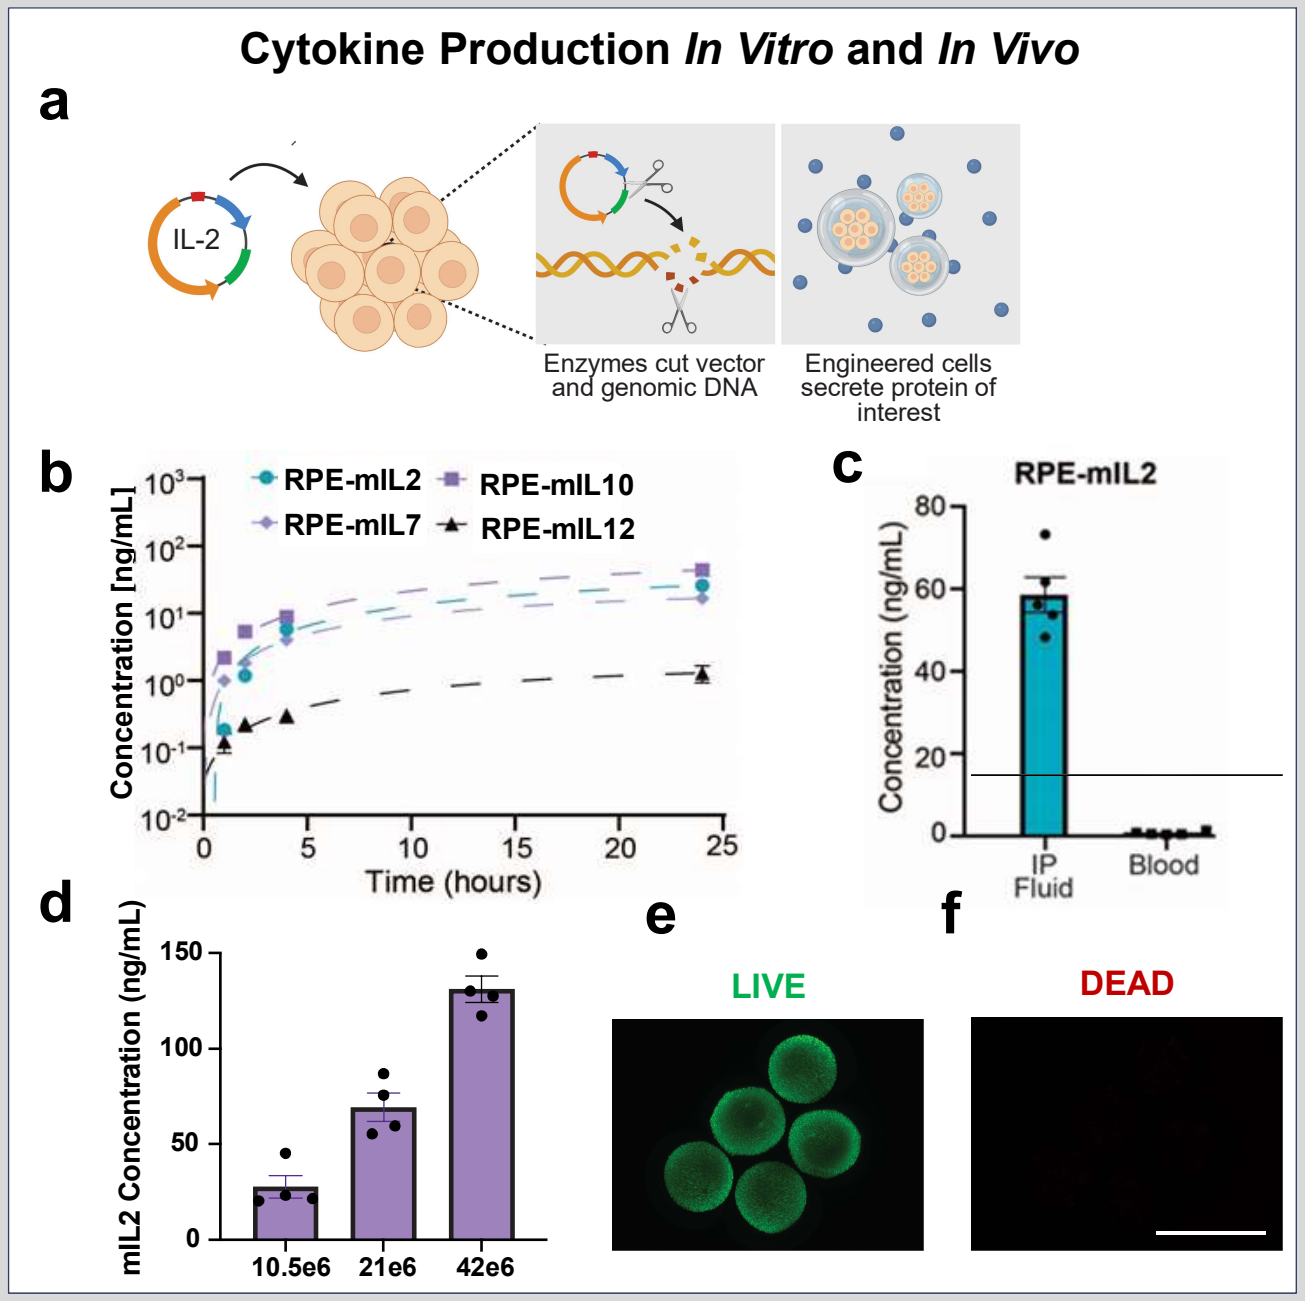
Cytokine Production In Vitro and In Vivo
a
IL-2
Enzymes cut vector and genomic DNA
Engineered cells secrete protein of interest
b
RPE-mIL2
RPE-mIL10
RPE-mIL7
RPE-mIL12
Concentration [ng/mL]
Time (hours)
c
RPE-mIL2
Concentration (ng/mL)
IP Fluid
Blood
d
mIL2 Concentration (ng/mL)
10.5e6
21e6
42e6
e
LIVE
f
DEAD

Above: a) Schematic demonstrating cell engineering and encapsulation. b) *In vitro* cytokine concentration at 1, 2, 4 and 24 hours (n=5). c) IL-2 concentration in IP fluid and blood of mice 24 hours after implantation (n=5). d) IL-2 concentration at 24 hours at various cell doses (n=4). e) Cells stained for viability after encapsulation. Calcein AM (Live), Ethidium homodimer-1 (dead), scale 2mm.

RPE-mIL2 monotherapy reduces MPM tumors and is well tolerated in mice

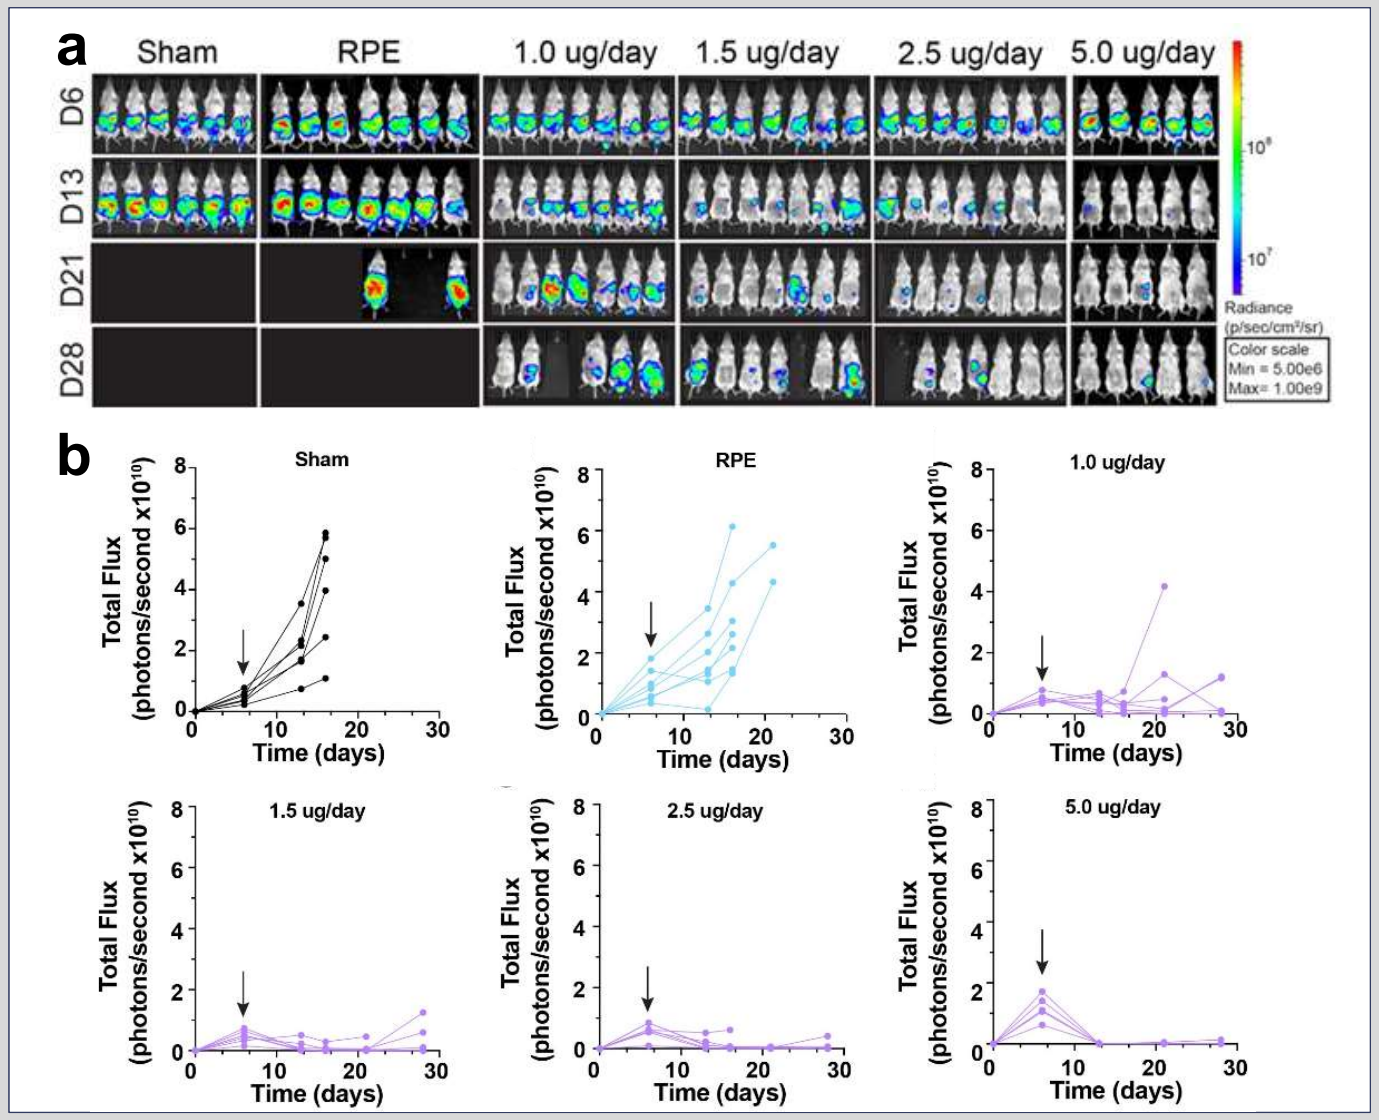
a
Sham
RPE
1.0 ug/day
1.5 ug/day
2.5 ug/day
5.0 ug/day
D6
D13
D21
D28
Radiance (p/sec/cm²/sr)
Color scale Min = 5.00e6 Max= 1.00e9
b
Sham
RPE
1.0 ug/day
1.5 ug/day
2.5 ug/day
5.0 ug/day
Total Flux (photons/second x10^10)
Time (days)

Above: a) IVIS images of mice from each treatment group over time following administration of sham surgery, RPE only, 1.0, 1.5, 2.5, or 5.0 ug of IL-2/day via RPE-mIL2 b) Total flux calculated from IVIS imaging over time after treatment (n=5-7).

RPE-mIL2+aPD-1 treatment eradicates MPM tumors in mice

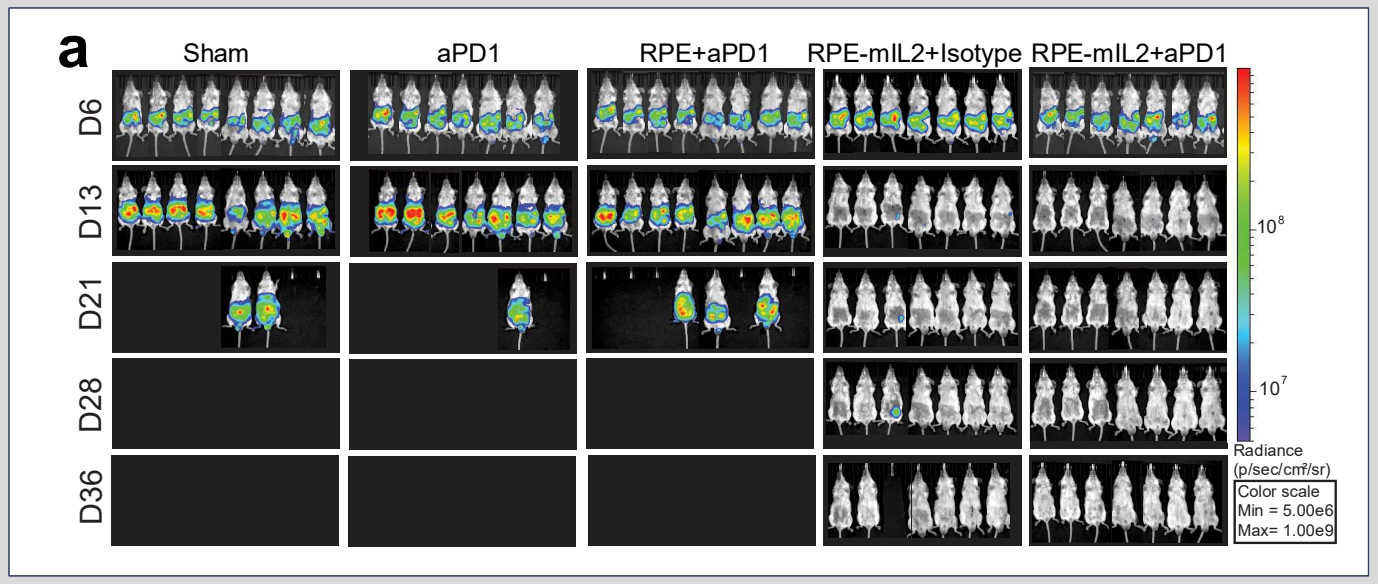
a
Sham
aPD1
RPE+aPD1
RPE-mIL2+Isotype
RPE-mIL2+aPD1
D6
D13
D21
D28
D36
Radiance (p/sec/cm²/sr)
Color scale Min = 5.00e6 Max= 1.00e9

Above: a) IVIS images of mice from each treatment group over time following administration of sham surgery, aPD1 injections, RPE+aPD1, RPE-mIL2+isotype, or RPE-mIL2 +aPD1 (n=7-8)

RPE-mIL2+aPD-1 treatment leads to protection from MPM recurrence in mice

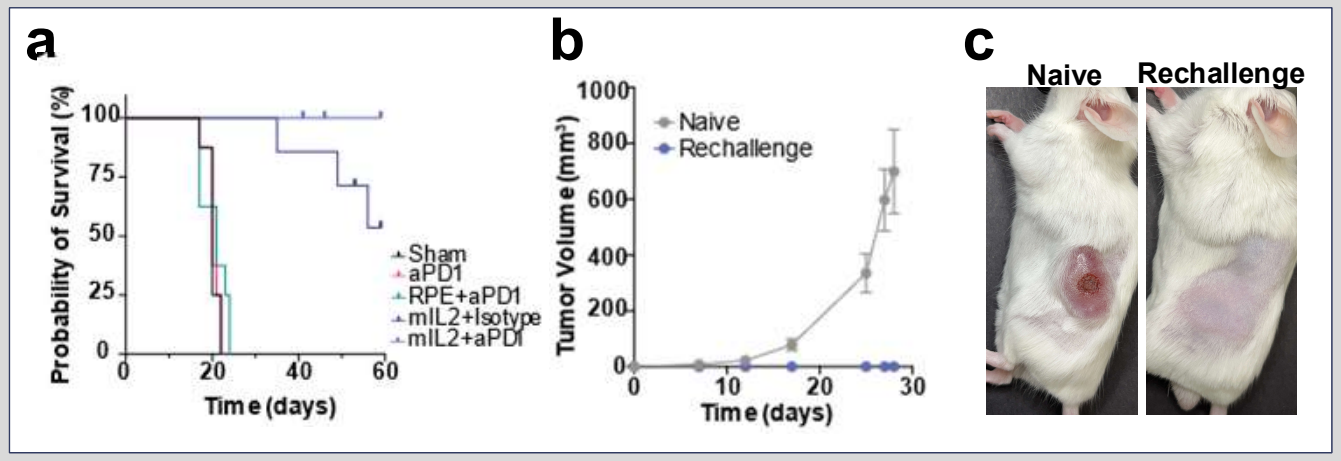
a
Probability of Survival (%)
Sham
aPD1
RPE+aPD1
mIL2+Isotype
mIL2+aPD1
Time (days)
b
Tumor Volume (mm³)
Naive
Rechallenge
Time (days)
c
Naive
Rechallenge

Above: Continuation of combination study. a) Survival curve of mice in each treatment group (n=7-8). b) Tumor volume measuring subcutaneous AB1 tumors in naïve or rechallenged mice (n=4-6). c) Representative images of subcutaneous tumor burden in naïve (left) or rechallenged (right) mice 28 days post rechallenge.

RPE-hIL2 is Well Tolerated in Primates

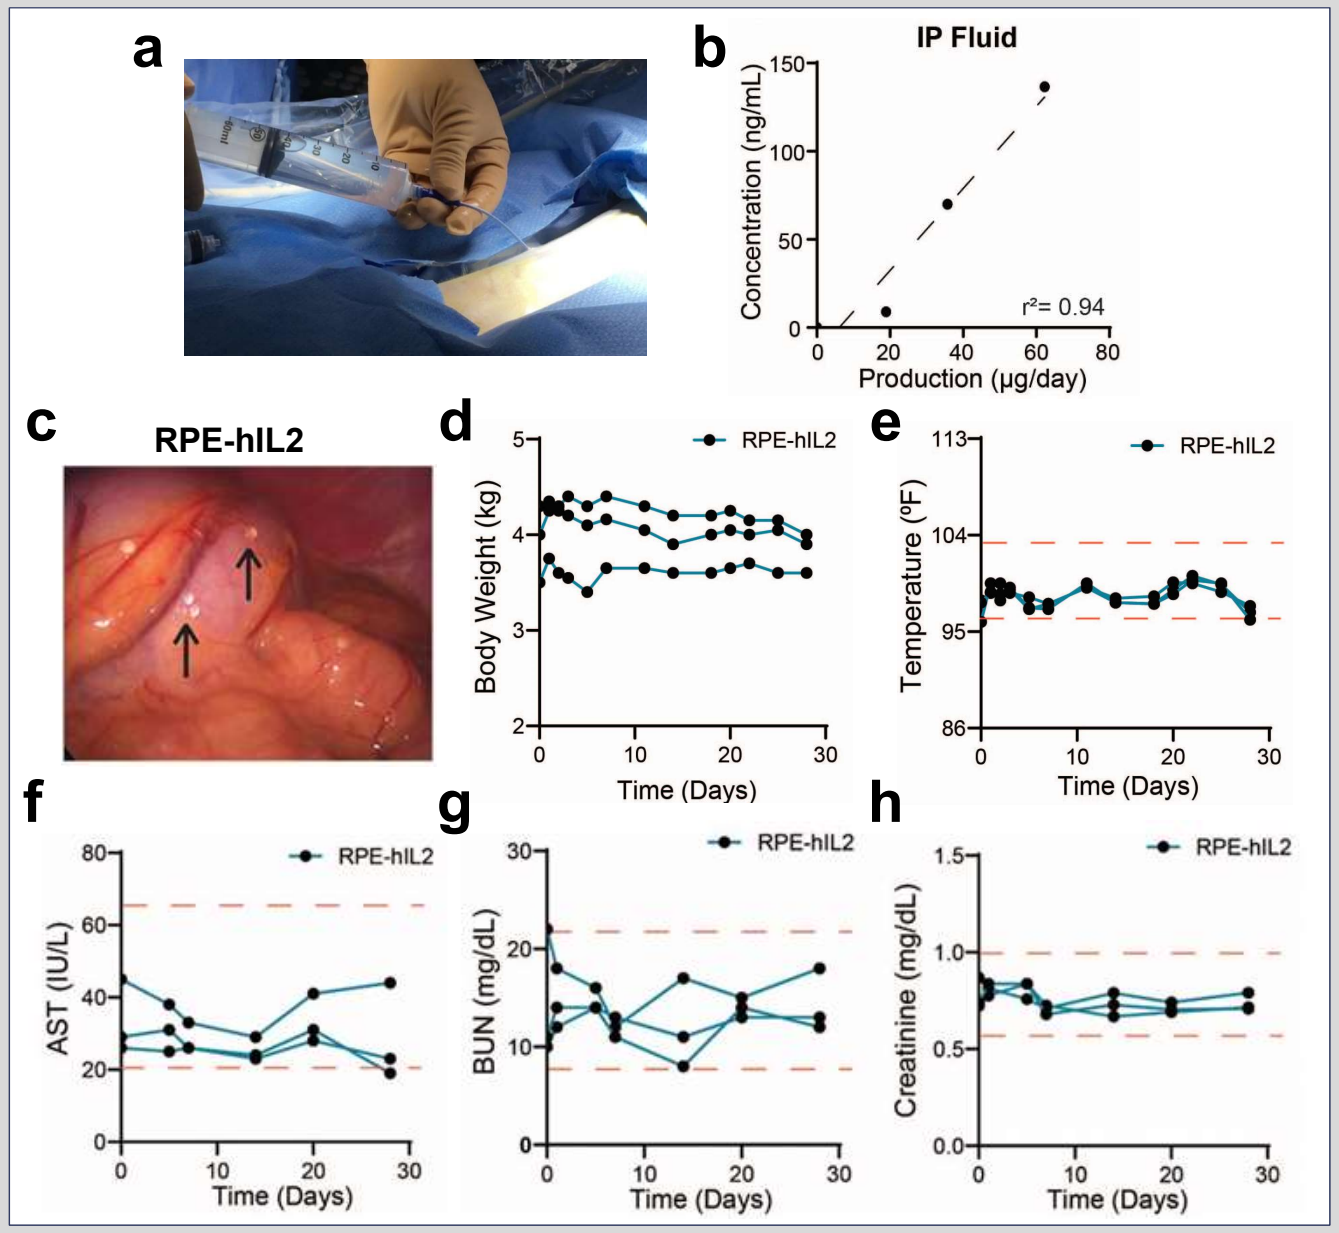
a
b
IP Fluid
Concentration (ng/mL)
r²= 0.94
Production (µg/day)
c
RPE-hIL2
d
RPE-hIL2
Body Weight (kg)
Time (Days)
e
RPE-hIL2
Temperature (ºF)
Time (Days)
f
RPE-hIL2
AST (IU/L)
Time (Days)
g
RPE-hIL2
BUN (mg/dL)
Time (Days)
h
RPE-hIL2
Creatinine (mg/dL)
Time (Days)

Above: a) Image of administration in primates. b) hIL2 concentration in IP fluid of primates. c) Laparoscopic images of RPE-hIL2 in the IP space of a cynomolgus macaque. Black arrows point to the cytokine factories. d) Body weight and e) temperature over time (n=3). f-h) AST, BUN, and creatinine level over time (n=3).

ACKNOWLEDGEMENTS

- Rice University Shared Equipment Authority
- Baylor University Cell Sorting Core
- MD Anderson Cancer Center
- Veiseh Lab - Rice University
- Peng Lab – University of Houston
- Funding support from Avenge Bio, Inc.