Tumor Adjacent Cytokine Factories for Eradication of Ovarian Cancer Tumor Burden in Mice Through Cytotoxic T-cell Activation with Safe and Predictable Dosing in Non-human Primates

Amanda M. Nash[1], Maria I. Jarvis[1], Samira Aghlara-Fotovat[1], Sudip Mukherjee[1], Andrea Hernandez[1], Andrew D. Hecht[1], Peter D. Rios[2], Sofia Ghani[2], Ira Joshi[2], Douglas Isa[2], Yufei Cui[1], Shirin Nouraein[1], Jared Z. Lee[1], Chunyu Xu[3], David Y. Zhang[1], Rahul A. Sheth[4], Weiyi Peng[3], Jose Oberholzer[2], Oleg A. Igoshin[1], Amir A. Jazaeri[5], and Omid Veiseh[1,#]

1. Department of Bioengineering, Rice University Houston, TX, 2. CellTrans, Inc., Chicago, IL, 3. Department of Biology and Biochemistry, The University of Houston, Houston, TX, 4. Department of Interventional Radiology, The University of Texas MD Anderson Cancer Center, Houston, TX, 5. Department of Gynecologic Oncology and Reproductive Medicine, Division of Surgery, The University of Texas MD Anderson Cancer Center, Houston, TX

Abstract

Pro-inflammatory cytokines have been approved by the FDA for the treatment of metastatic melanoma and renal carcinoma. However, effective cytokine therapy is limited by its short half-life in circulation and the severe adverse effects associated with high systemic exposure. To overcome these limitations, we developed a clinically translatable localized cytokine delivery platform composed of polymer encapsulated epithelial cells that produce localized natural cytokines (IL2, IL7, IL10, or IL12) with temporal regulation. Tumor-adjacent local administration of these cytokine factories demonstrated predictable dose modulation with spatial and temporal control and provided ovarian cancer immunotherapy without systemic toxicities. Treatment of peritoneal tumors using IL2 producing cytokine factories provided sustained eradication of peritoneal tumors in an ovarian cancer mouse model. Our data confirmed local increases in the activation (CD25+CD8+) and proliferation (Ki67+CD8+) of cytotoxic T cells within the IP space of cytokine factory treated mice. Significantly, this platform produced local and systemic T cell biomarker profiles that predict efficacy without toxicity in non-human primates.

Localized Dose for Personalized Treatment

Cytokine Production *In Vitro* and *In Vivo*

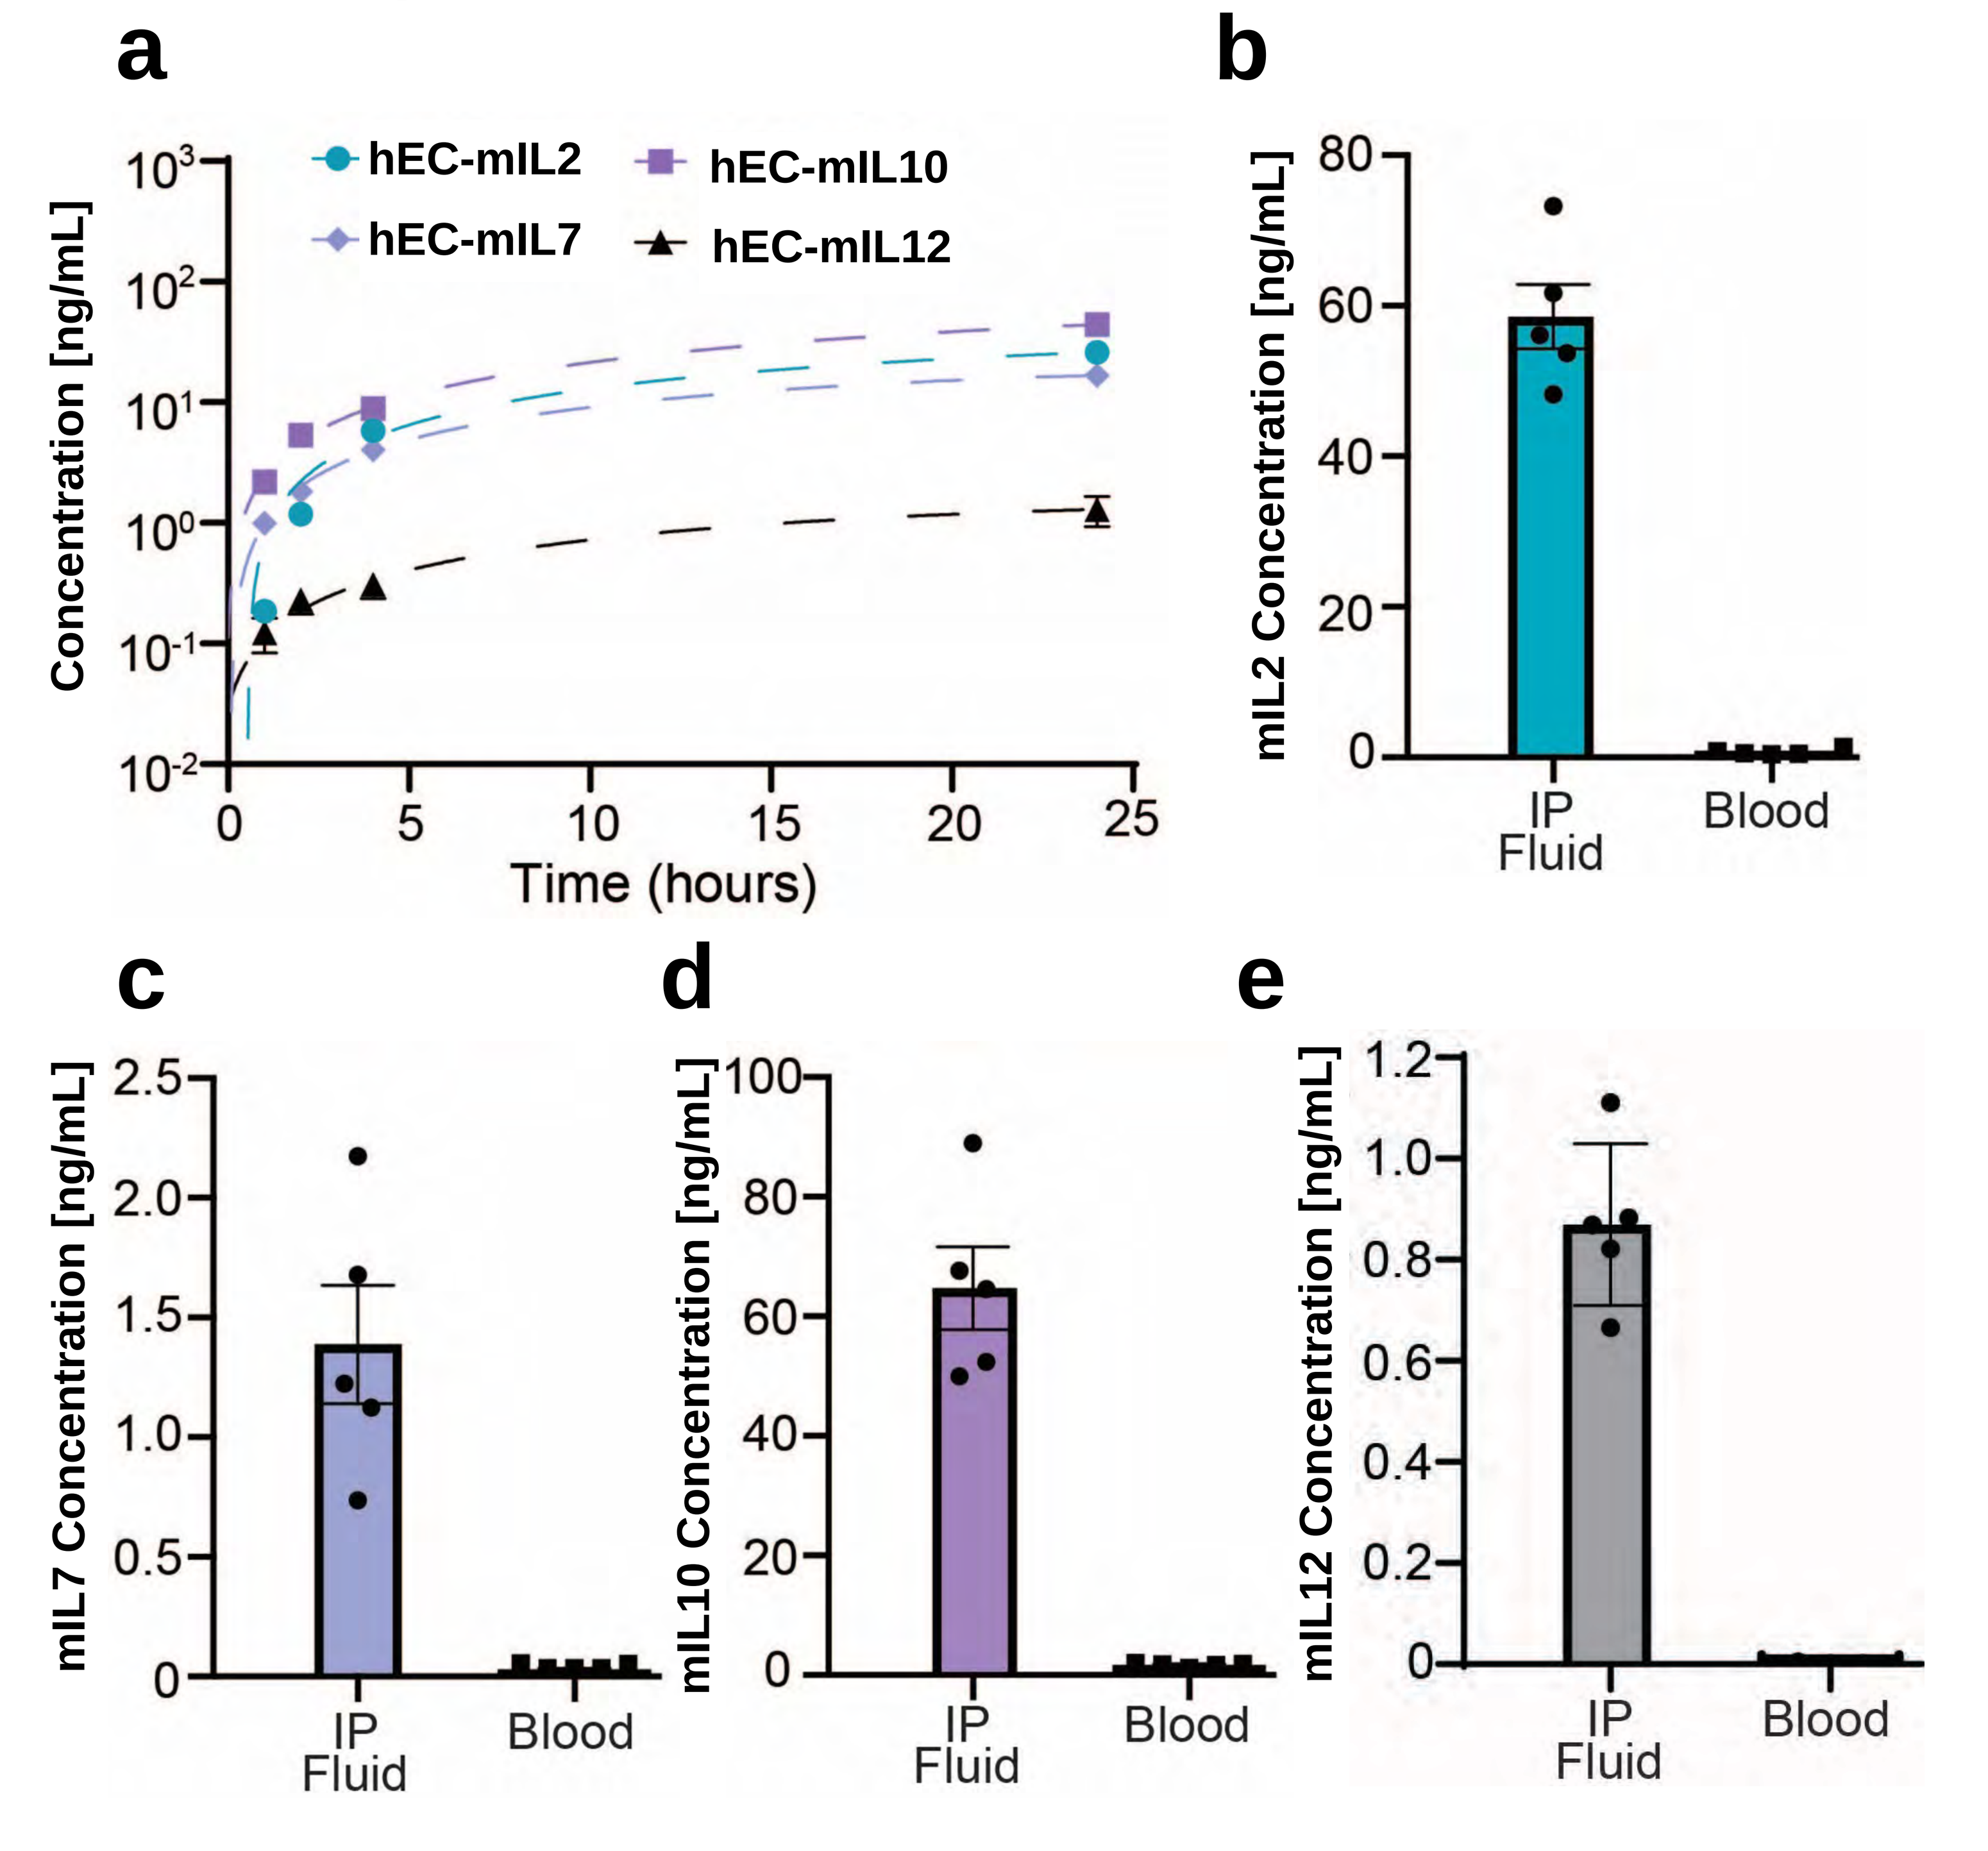
a
hEC-mIL2
hEC-mIL10
hEC-mIL7
hEC-mIL12
Concentration [ng/mL]
Time (hours)
b
mIL2 Concentration [ng/mL]
IP Fluid
Blood
c
mIL7 Concentration [ng/mL]
d
mIL10 Concentration [ng/mL]
e
mIL12 Concentration [ng/mL]

Above: a) *In vitro* cytokine concentration at 1, 2, 4 and 24 hours (n=5). b-e) Cytokine concentration in IP fluid of mice 24 hours after implantation of hEC-mIL2, hEC-mIL7, hEC-mIL10, or hEC-mIL12 (n=5). Concentrations were measured using ELISA.

hEC-mIL2 Monotherapy Eradicates ID8/MOSEC Ovarian Cancer

ID8-Fluc Total Flux Over Time

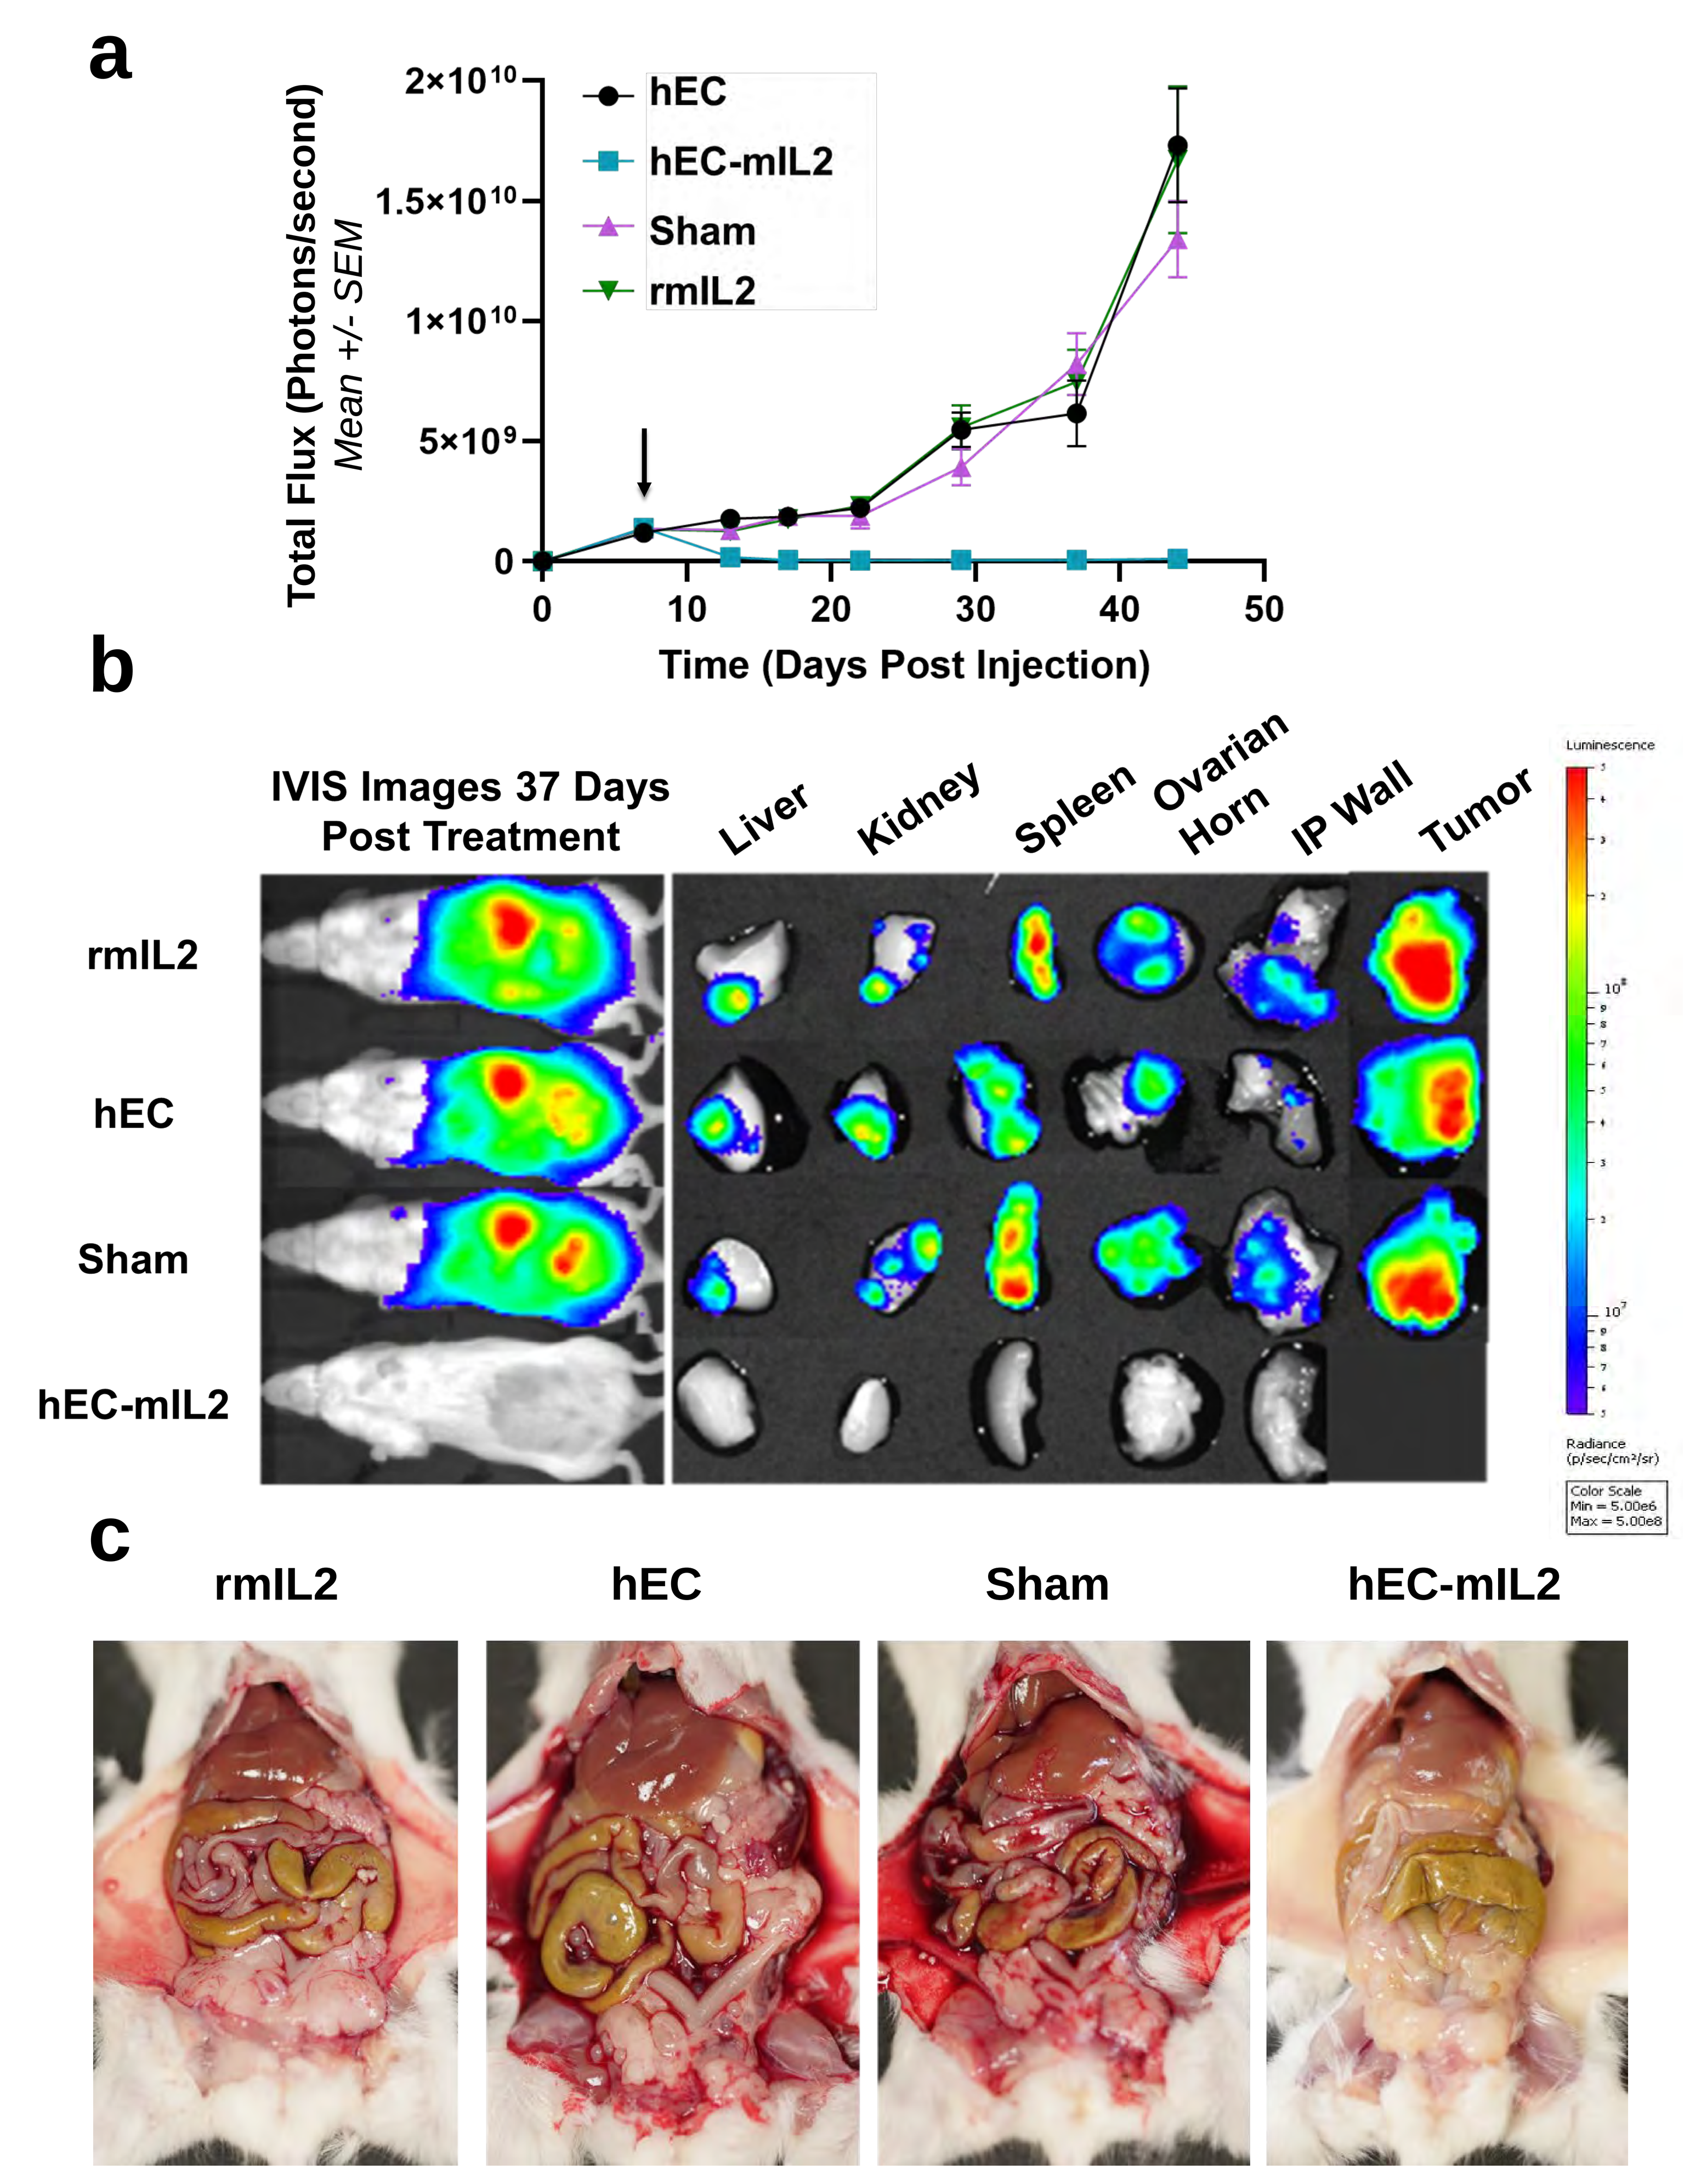
a
Total Flux (Photons/second)
Mean +/- SEM
hEC
hEC-mIL2
Sham
rmIL2
Time (Days Post Injection)
b
IVIS Images 37 Days Post Treatment
Liver
Kidney
Spleen
Ovarian Horn
IP Wall
Tumor
rmIL2
hEC
Sham
hEC-mIL2
c
rmIL2
hEC
Sham
hEC-mIL2

Above: a) Total flux calculated from IVIS imaging over time after treatment with hEC-mIL2, hEC only, recombinant IL2 injections or sham surgery (n=5). Black arrow represents total flux at time of treatment. b) (left) Representative IVIS images of each treatment group 37 days post treatment. (right) Representative IVIS images of ex vivo organs immediately after sacrifice. c) Representative images of intraperitoneal space of each treatment group during necropsy.

hEC-mIL2 Increases Local Cytotoxic T cell Proliferation

Local T-cell Subsets 7 Days Post-Treatment

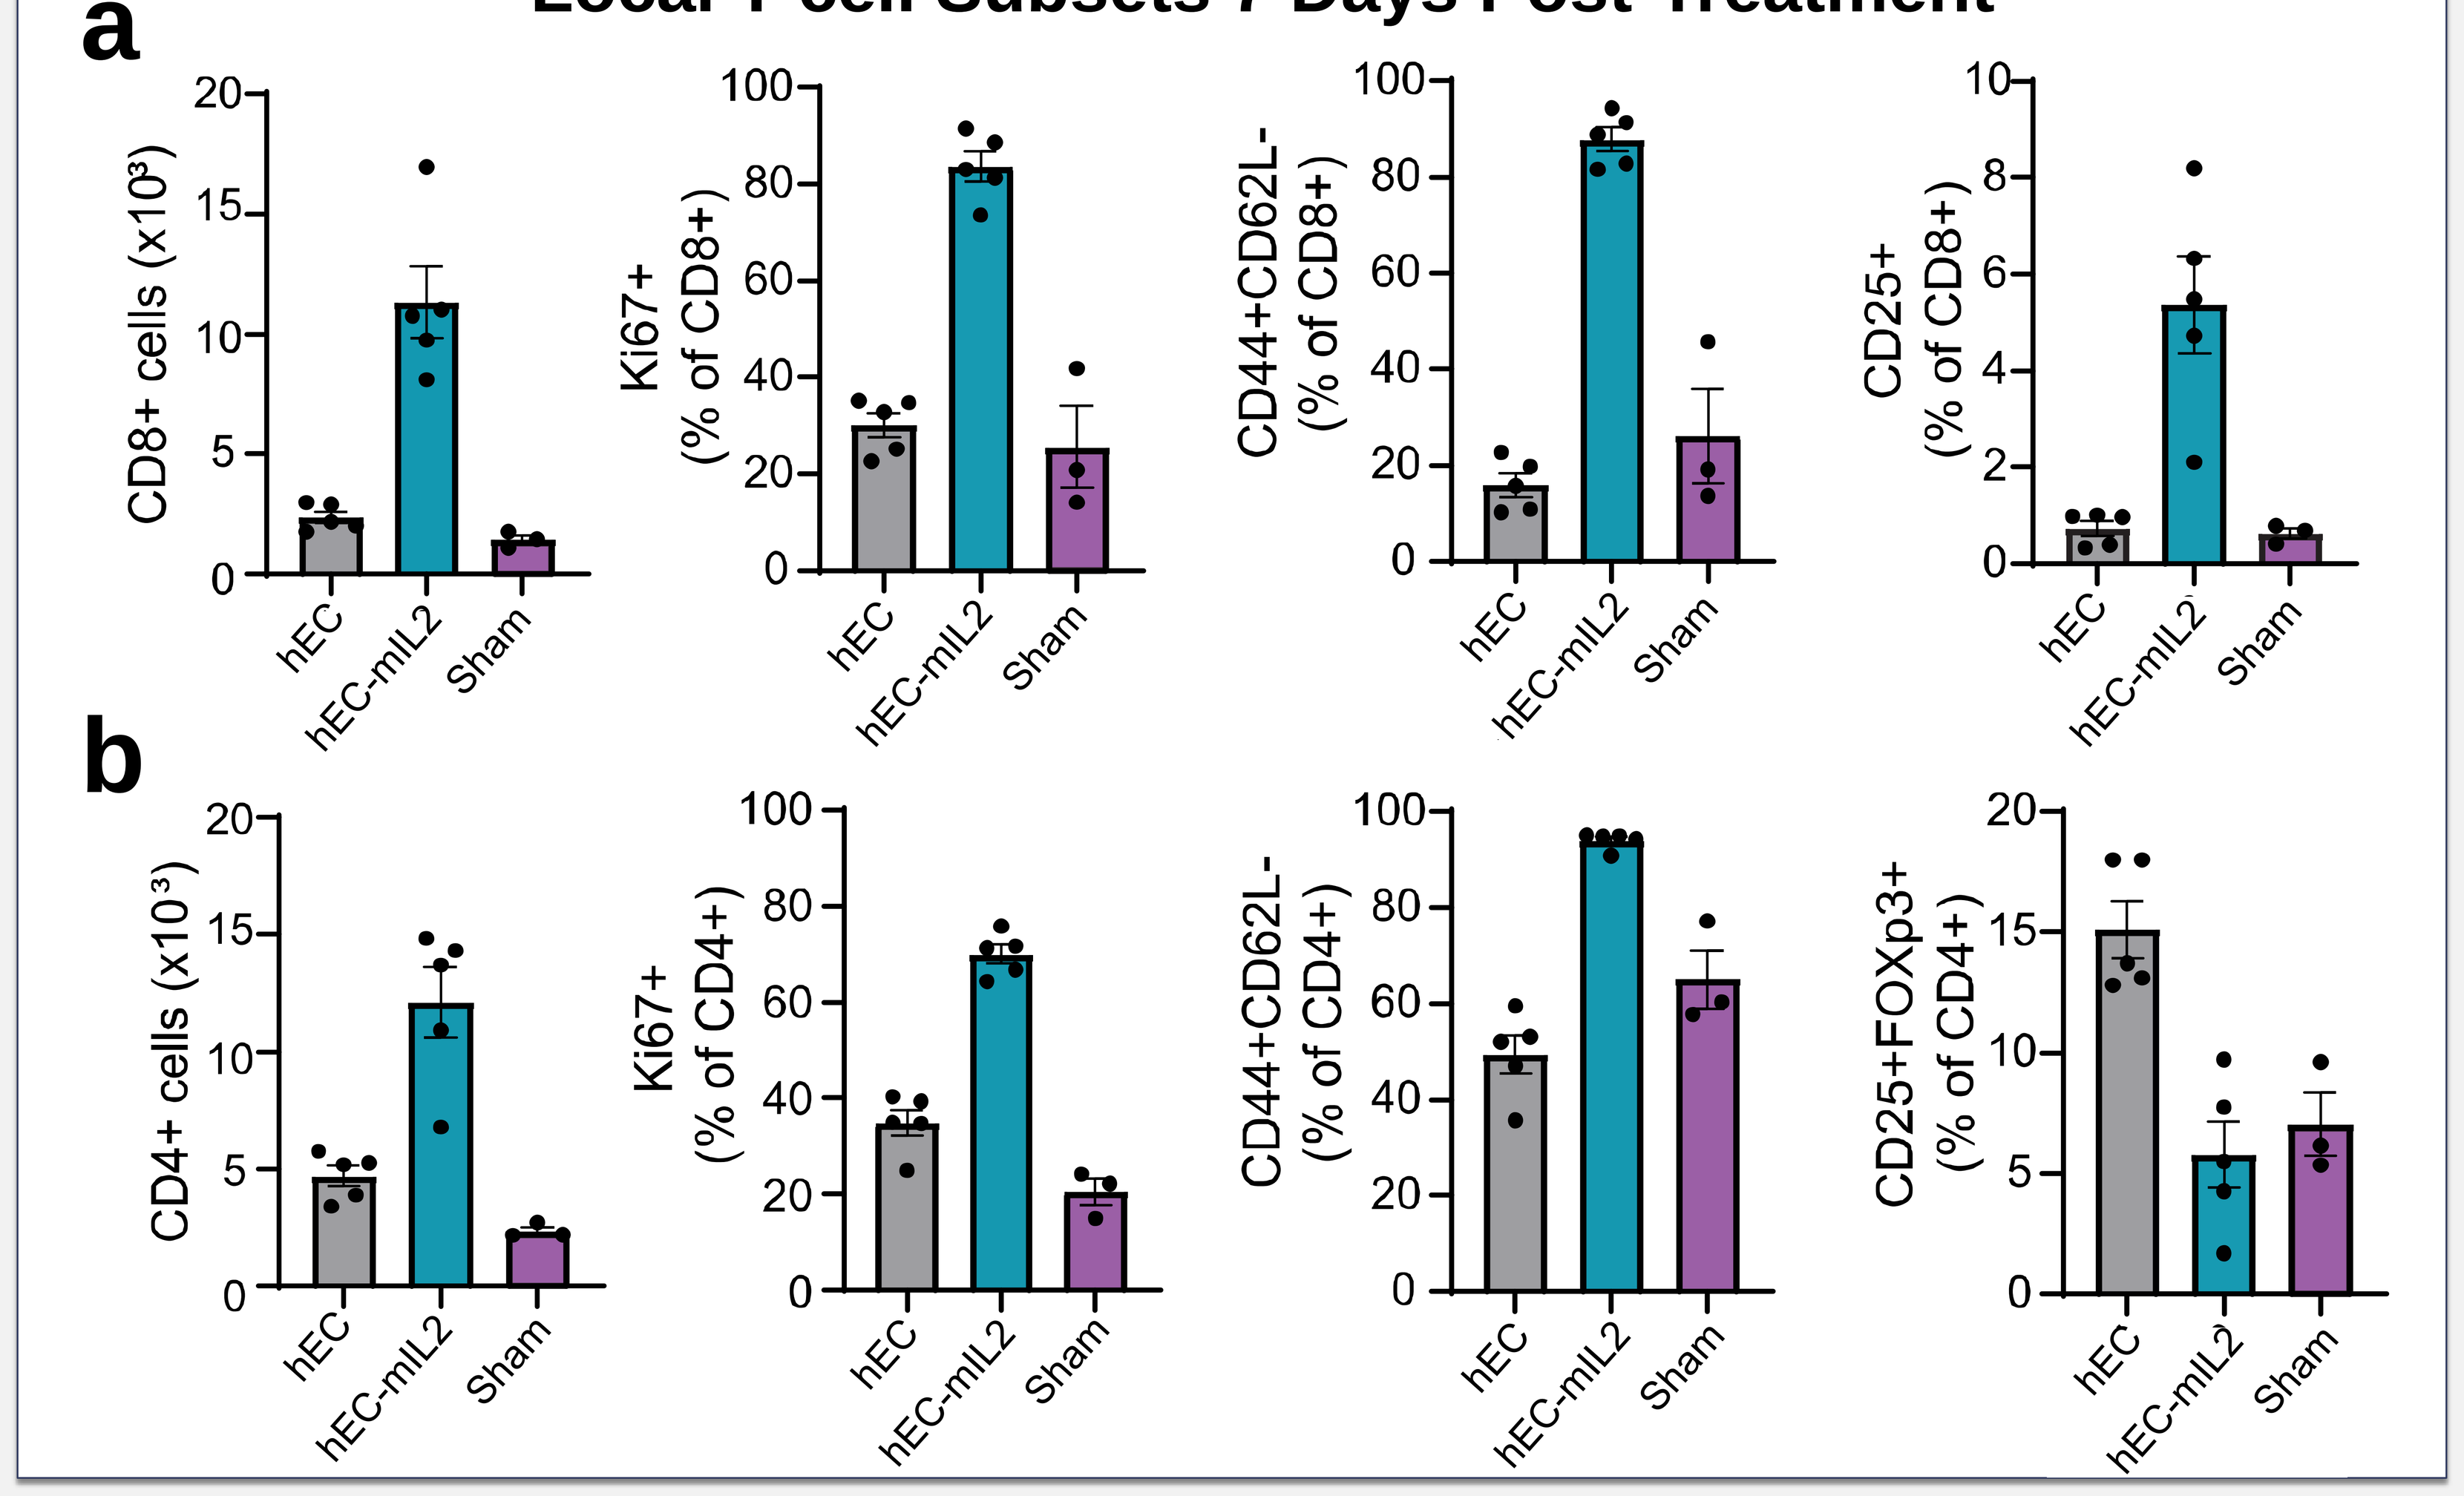
a
CD8+ cells (x10³)
Ki67+ (% of CD8+)
CD44+CD62L- (% of CD8+)
CD25+ (% of CD8+)
hEC
hEC-mIL2
Sham
b
CD4+ cells (x10³)
Ki67+ (% of CD4+)
CD44+CD62L- (% of CD4+)
CD25+FOXp3+ (% of CD4+)

Above: a) CD8+ counts, percentage of Ki67+, CD25+, or CD44+CD62L- T cells as frequency of CD8+ T cells, b) CD4+ counts, percentage of Ki67+, CD25+, or CD44+CD62L- T cells as frequency of CD4+ T cells analyzed via flow cytometry.

hEC-hIL2 is Well Tolerated in Primates

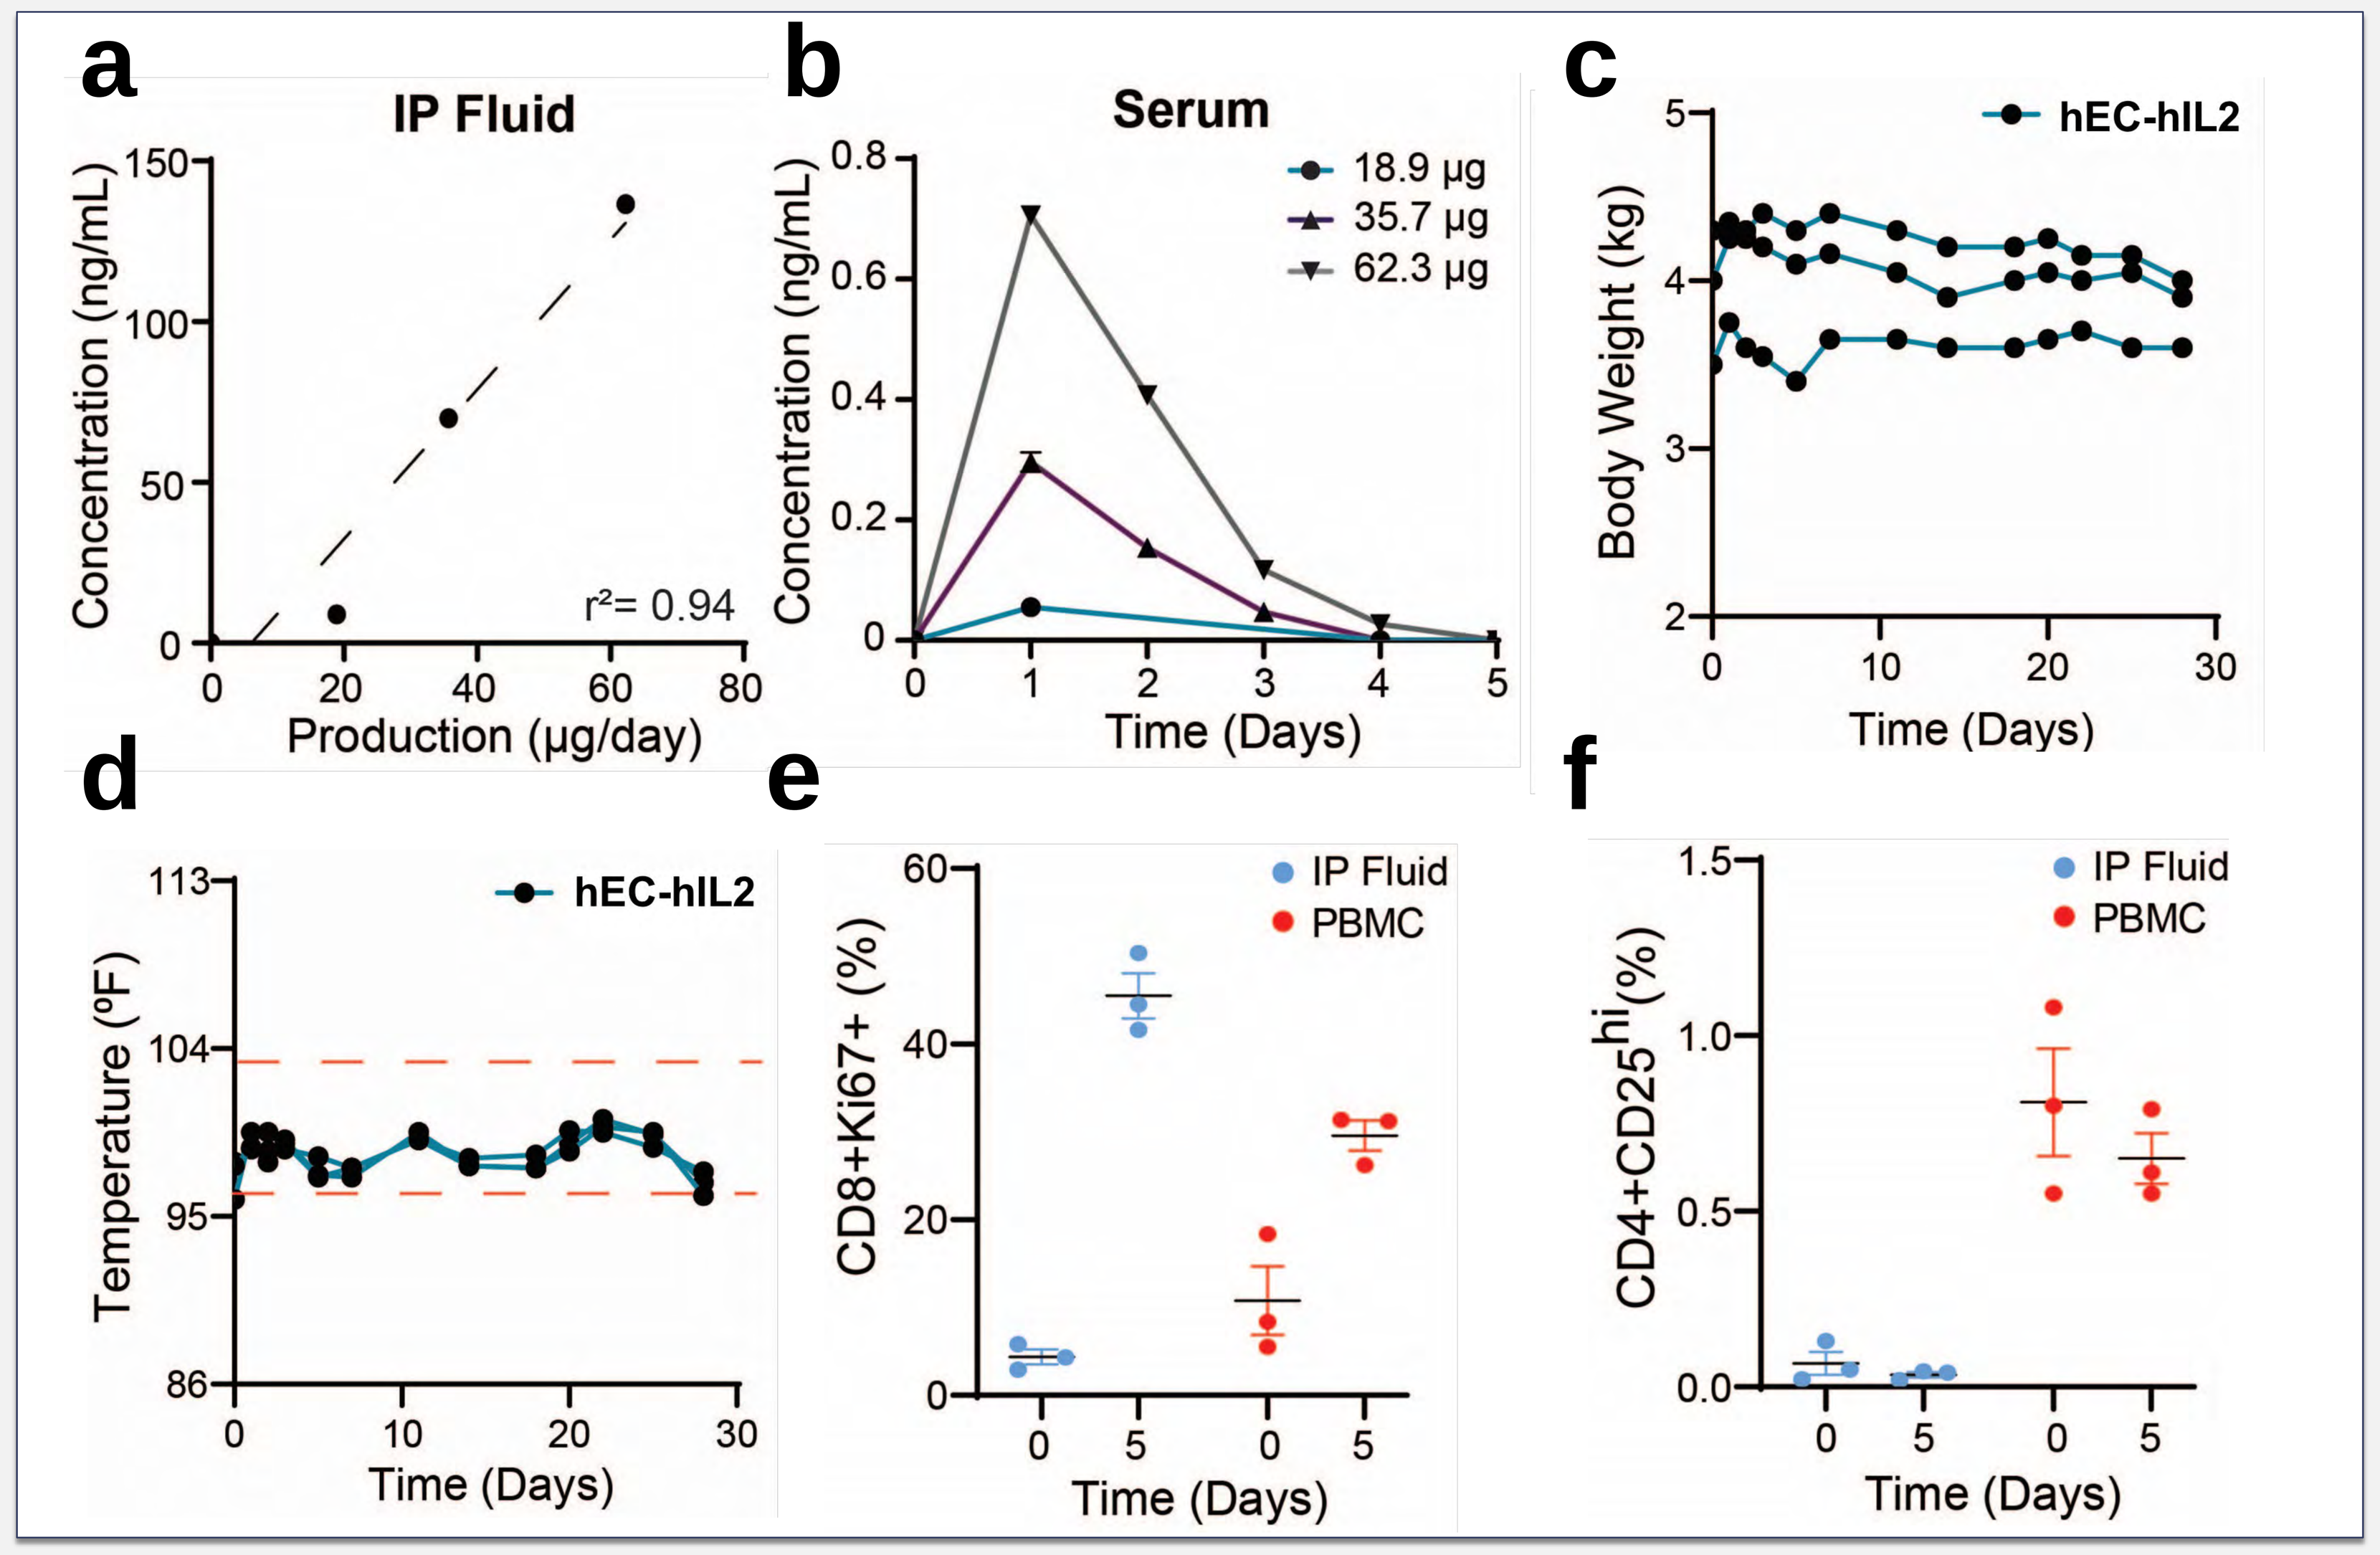
a
IP Fluid
Concentration (ng/mL)
Production (µg/day)
r²= 0.94
b
Serum
18.9 µg
35.7 µg
62.3 µg
Time (Days)
c
hEC-hIL2
Body Weight (kg)
d
Temperature (ºF)
e
CD8+Ki67+ (%)
IP Fluid
PBMC
f
CD4+CD25hi (%)

Above: a) IP hIL2 concentration as a function of dose 5 days post administration. b) Serum hIL2 concentration over time. c) Body weight and d) temperature over time (n=3). e) Percentage of Ki67+ cells as a frequency of CD8+ T cells. f) Percentage of regulatory T cells as a frequency of total cells.

Acknowledgements

- Rice University Shared Equipment Authority
- Baylor University Cell Sorting Core
- MD Anderson Cancer Center

•Veiseh Lab - Rice University
•Peng Lab – University of Houston
•Funding support from Avenge Bio, Inc.

National Institutes of Health